Aseptically Processed Cryopreserved Amnion Membrane Preserves Essential Bioactive Components that Support the Progression of the Wound Healing Cascade

[1]Dasgupta, A; [1]Wu, K; [1]Madans, A; [1]Chnari, E; [1]Jacobs, M

[1]Musculoskeletal Transplant Foundation (MTF), Edison, NJ

INTRODUCTION

The normal wound healing cascade consists of three overlapping phases: inflammation, proliferation and remodeling [1,2]. However, in chronic wounds this process can be overwhelmed by the inflammation phase due to an imbalance, preventing advancement to the proliferation phase and leaving the wound unable to heal. There are a number of well documented key biological components that work together at each phase and participate in successfully moving the wound healing cascade forward [2–4].

Amnion membranes have been employed in the treatment of wounds since the 1910s [5,6]. They are rich in matrix proteins, growth factors and contain stem cells capable of multilineage differentiation as well as growth factor production and secretion [7–12]. The matrix proteins maintain the integrity and function of the basement membrane, aid in angiogenesis and support cell attachment during wound healing. The plethora of growth factors/cytokines supports the healing process to improve wound closure and reduce scar formation. Aseptically processing fresh amnion tissue and cryopreserving the membrane can preserve these inherent wound healing attributes, thereby providing a natural matrix scaffold, inherent growth factors and cytokines along with preserving viable, non-immunogenic cells.

MATERIALS AND METHODS

Fresh amnion membranes were processed aseptically and cryopreserved, without terminal sterilization, at the Musculoskeletal Transplant Foundation (MTF, Edison NJ).

Cryopreserved amnion membranes were thawed and then characterized via immuno-histochemical staining for matrix proteins, growth factors and anti-inflammatory cytokines.

Cell viability was characterized by live/dead cell staining (LIVE/DEAD Viability/Cytotoxicity Kit, Invitrogen) and labeled cells were enumerated via fluorescent microscopy. In addition, cell functionality of the viable cells was determined by tissue explant cultures to observe cell migration and cell proliferation. Mixed lymphocyte reaction testing (Marin Biologics, CA) was performed to assess cell non-immunogenicity.

RESULTS

Immunohistochemical staining revealed the retention of various matrix proteins such as collagen I, III, VI, VII, nidogen-1 and fibronectin. Additionally, positive staining demonstrated the preservation of wound healing growth factors including PDGF-AA, PDGF-BB, VEGF, EGF, FGF-2, TGF-b1 and anti-inflammatory cytokines such as IL-6 and IL-10 as well as antimicrobial peptide ß-defensin-1 in the cryopreserved membrane.

Viability analysis showcased greater than 70% cell viability on cryopreserved and thawed membranes and tissue explant cultures demonstrated cell migration and proliferation from the membrane onto the well plate surface. Immunogenicity testing of the amnion membrane was evaluated via mixed lymphocyte reaction, revealing that the cells present in the allograft were non-immunogenic.

© 2015 Musculoskeletal Transplant Foundation 4/15 00049 Mktg -1113 Rev. 0

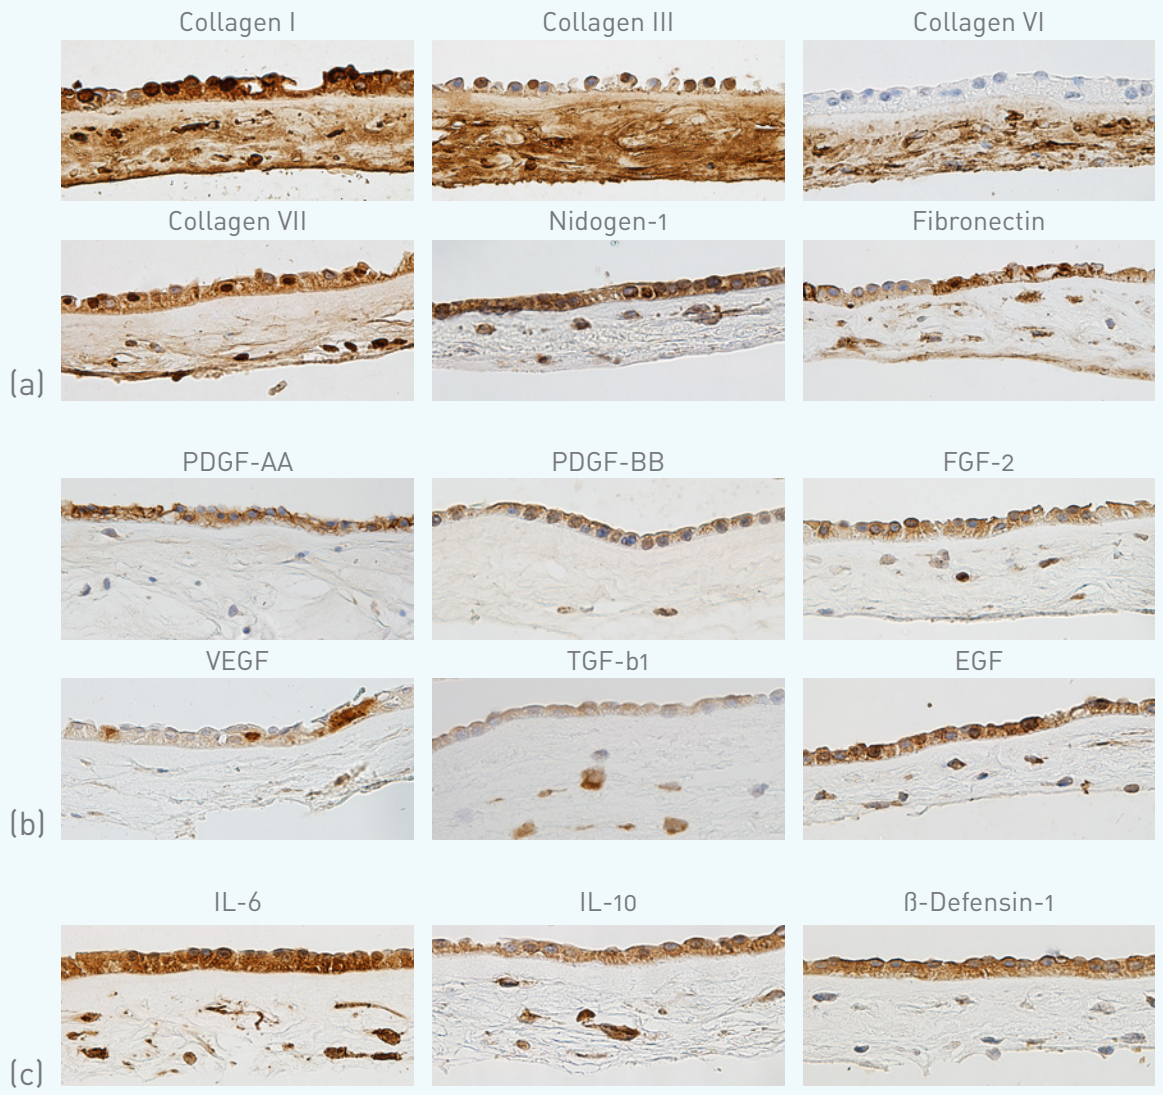
Collagen I
Collagen III
Collagen VI
Collagen VII
Nidogen-1
Fibronectin
(a)
PDGF-AA
PDGF-BB
FGF-2
VEGF
TGF-b1
EGF
(b)
IL-6
IL-10
ß-Defensin-1
(c)

Figure 1.(a) Immunohistochemical (IHC) staining of cryopreserved amnion membrane for matrix proteins shows retention of collagens I, III, VI, VII as well as nidogen-1 and fibronectin; (b) IHC staining for growth factors demonstrates presence of PDGF-AA, PDGF-BB, FGF-2, VEGF, TGF-ß1, and EGF; (c) IHC staining for cytokines demonstrates preservation of IL-6, IL-10, and ß-defensin-1 in the cryopreserved membrane.

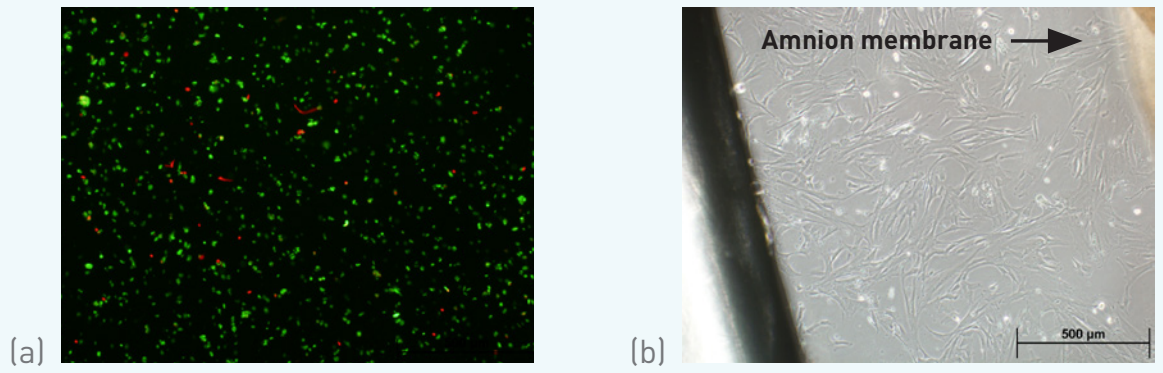
(a)
(b)
Amnion membrane
500 µm

Figure 2. (a) Cells were enzymatically removed from the cryopreserved amnion membrane and then stained using a live/dead kit. Under fluorescent microscopy, cells with intracellular esterase activity, an indicator of metabolic activity and viability, appear green while non-viable cells with compromised plasma membranes appear red. (b) Explant cultures of cryopreserved amnion membrane showed cells migrating from the tissue onto the well plate surface and proliferating, indicating activity and functionality of the viable cells.

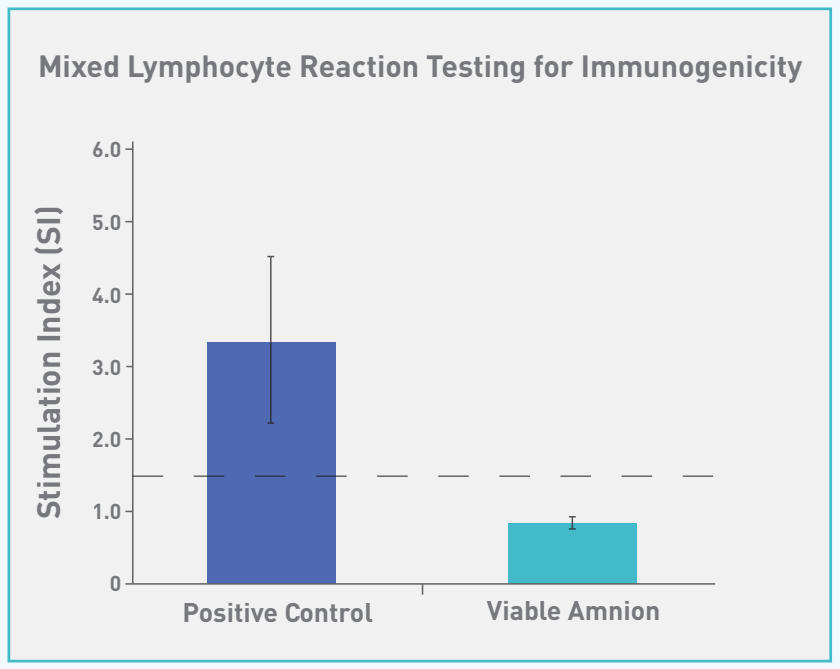
Mixed Lymphocyte Reaction Testing for Immunogenicity
Stimulation Index (SI)
6.0
5.0
4.0
3.0
2.0
1.0
0
Positive Control
Viable Amnion

Figure 3. Mixed lymphocyte reaction testing performed on cryopreserved viable amnion membrane shows no immunogenic reaction as compared to positive control and is below the index threshold value considered "immunogenic".

CONCLUSION

The data demonstrates that aseptic fresh tissue processing and cryopreservation retains critical wound healing biological components in the amnion membrane. The synergistic interplay of the inherent matrix proteins, growth factors, and viable cells can support the progression of the wound healing cascade.

REFERENCES

1. Sinno H, Prakash S. Complements and the Wound Healing Cascade: An Up-dated Review. *Plastic Surgery International.* 2013;2013:146764.
2. Demidova-Rice TN, Hamblin MR, Herman IM. Acute and Impaired Wound Healing: Pathophysiology and Current Methods for Drug Delivery, Part 1: Normal and Chronic Wounds: Biology, Causes, and Approaches to Care. *Advances in Skin & Wound Care.* 2012;25(7):304-314.
3. Werner S, Grose R. Regulation of wound healing by growth factors and cyto-kines. *Physiol Rev.* 2003 Jul;83(3):835-70.
4. Cross KJ, Mustoe TA. Growth factors in wound healing. *Surg Clin North Am.* 2003 Jun;83(3):531-45, vi.
5. Davis J. Skin Transplantation: With a review of 550 cases at John Hopkins Hospital. *John Hopkins Hosp Rep.* 1910;15:310.
6. Sabella N. Use of fetal membranes in skin grafting. *Med Records NY.* 1913; 83:478-480.
7. Bryant-Greenwood GD. The extracellular matrix of the human fetal mem-branes: structure and function. *Placenta.* 1998 Jan;19(1):1-11.
8. Díaz-Prado S, Muiños-López E, Hermida-Gómez T, Cicione C, Rendal-Vázquez ME, Fuentes-Boquete I, de Toro FJ, Blanco FJ. Human amniotic membrane as an alternative source of stem cells for regenerative medicine. *Differentiation.* 2011 Mar;81(3):162-71. doi: 10.1016/j.diff.2011.01.005.
9. Ilancheran S, Michalska A, Peh G, Wallace EM, Pera M, Manuelpillai U. Stem cells derived from human fetal membranes display multilineage differentiation potential. *Biol Reprod.* 2007 Sep;77(3):577-88.
10. Koizumi NJ, Inatomi TJ, Sotozono CJ, Fullwood NJ, Quantock AJ, Kinoshita S. Growth factor mRNA and protein in preserved human amniotic membrane. *Curr Eye Res.* 2000 Mar;20(3):173-7.
11. Niknejad H, et al. Properties of the Amniotic Membrane for Potential Use in Tissue Engineering. *European Cells and Materials.* 2008. 15:88-99.
12. Wolbank S, Hildner F, Redl H, van Griensven M, Gabriel C, Hennerbichler S. Impact of human amniotic membrane preparation on release of angiogenic factors. *J Tissue Eng Regen Med.* 2009 Dec;3(8):651-654.